1025

African Ethnicity Is Associated with Slower Disease Progression in the Swiss HIV Cohort Study (SHCS)

Viktor Müller[1,7], Viktor von Wyl[2], Sabine Yerly[3], Jürg Böni[4], Thomas Klimkait[5], Philippe Bürgisser[6], Bruno Ledergerber[2], Huldrych F. Günthard[2], Sebastian Bonhoeffer[7] and the SHCS

1) Eötvös Loránd University, Budapest, Hungary; 2) University Hospital Zürich, Zürich, Switzerland; 3) Geneva University Hospital, Geneva, Switzerland; 4) University of Zürich, Zürich, Switzerland; 5) University of Basel, Basel, Switzerland; 6) Lausanne University Hospital, Lausanne, Switzerland; 7) Federal Institute of Technology (ETH) Zürich, Zürich, Switzerland

muller_v@ludens.elte.hu
Tel:+3612090555/8798
Fax:+3613812188
ELTE Institute of Biology
Pázmány P. s. 1/C
1117 Budapest, Hungary

Abstract

Background: The rate of disease progression in HIV infection has been shown to be influenced by host factors, including ethnicity/descent. However, the effect of descent is often confounded by differences in the infecting viral subtypes and in socioeconomic/environmental factors; previous studies on the effect of African vs. European descent have yielded conflicting results. We now investigated the effect in the Swiss HIV Cohort Study (SHCS) that has limited environmental variation, and we have also determined viral subtype to control for the influence of this factor.

Methods: We estimated the linear rate of decline of the CD4 count and the setpoint viral load in treatment-naïve patients of African or European descent in the SHCS. The effect of descent was assessed by multivariate regression models including gender, descent, viral subtype, the earliest date of confirmed infection, age and the baseline CD4+ cell count; we also performed matched comparisons between patients of African and European descent based on the baseline CD4 count.

Results: We found that the decline slope of the CD4 count was significantly less steep (+26.6 cells/µL/year; 95% CI: 12.3–41.0; p<0.001) in patients of African descent (n=123) compared with patients of European descent (n=463), and this effect was independent of differences in the infecting viral subtypes. Matched comparisons confirmed the effect of African descent (p<0.001). Remarkably, the rate of CD4 decline depended strongly on the viral setpoint in patients of European descent (-42.1 cells/µL/year/log10 RNA copies/mL; 95% CI: -53.0–-31.2; p<0.001), but not in patients of African descent. The potential confounding effect of subtype-dependent sensitivity in newer generation real time PCR detection systems could be ruled out based on a sensitivity analysis.

Conclusions: Slower disease progression in patients of African descent might be related to host factors allowing better tolerance of high virus levels in patients of African descent compared with patients of European descent. We hypothesize that this difference might reflect evolutionary adaptation to higher overall levels of antigenic exposure in Africa.

Methods

Study population/inclusion criteria:

Patients of African or European descent in the SHCS, with CDC Stage A at entry to the cohort and registered heterosexual exposure category. The majority of patients of African descent in the cohort were immigrants from Sub-Saharan Africa and belonged to the heterosexual exposure category.

Data selection:

We used CD4 count and virus load data preceding the first initiation of antiretroviral treatment, and we discarded data points obtained during the first 200 days after the earliest date of confirmed infection and also data points after the first CD4 count below 100 cells/µL, to exclude the confounding effects of primary and late-stage infection [1].

Table 1. Demographic and clinical characteristics of the study group in the analysis of the CD4 slope .

Characteristic	Descent: African (n=123)		Descent: European (n=463)	
Female	91 (74%)		262 (56.6%)	
	Median	Interquartile range	Median	Interquartile range
ART-free follow-up (y)	4.2	2.9-5.8	5.0	3.5-7.9
Date of confirmed infection	17/08/01	17/01/99 – 18/12/03	21/04/97	14/12/90 – 27/10/01
Age at confirmed infection (y)	28.8	26.2 – 34.2	31.2	26.2 – 39.3
Baseline CD4+ cell count (cells/µL)	494	370.5 – 629.5	577	420 – 746
CD4 slope (cells/µL/year)	-28.2	-52.8 – -8.5	-52.5	-93.6 – -24.0
Setpoint (log10 RNA copies/mL)	3.72	3.19 – 4.35	4.02	3.28 – 4.51

Setpoint could be calculated for 103/242 patients of African/European descent from this group.

Markers of disease progression:

CD4 SLOPE was estimated by linear regression in patients with at least five CD4 counts spanning at least one year.
VIRAL SETPOINT was calculated as the mean log10 virus load per mL in the first two years of observations in patients with at least three data points.

Subtyping:

HIV-1 subtypes were defined using the REGA HIV-1 Subtyping Tool [2] based on HIV-1 pol sequences from the SHCS drug resistance database, which contains all genotypic HIV resistance tests performed by the four laboratories engaged in resistance testing in Switzerland, and which is stored in a central database developed and hosted by SmartGene (Zug, Switzerland, IDNS version 3.5.0). Sequences for which the REGA tool returned undetermined subtype were also analyzed with the STAR subtyping tool [3].

Statistical analyses:

REGRESSION ANALYSES (general linear models) were used to study the effect of potential explanatory variables on the CD4 slope and the viral setpoint. The primary explanatory variable of interest was descent (African vs. European), and other variables were included for adjustment purposes. Categorical factors included gender, descent and (in some analyses) viral subtype; reference levels were chosen as female, European and subtype B, to reflect the majority in the study group. Continuous predictors included the earliest date of confirmed infection (converted to Julian date), age at the date of confirmed infection and the baseline CD4+ cell count; setpoint was included as additional continuous predictor in some of the CD4 slope analyses. The starting models included all interaction terms between categorical predictors and two-way interactions between all pairs of a continuous and a categorical predictor. Significant effects were assessed by stepwise elimination of non-significant interaction terms and non-significant factors, and finally by merging non-significant levels of categorical predictors.

MATCHED COMPARISONS based on baseline CD4 count between patients of African and European descent were performed both with and without replacement. With replacement, the patient of European descent with the closest matching CD4 count was paired with each patient of African descent, allowing for multiple selection of individuals of European descent. Without replacement, each individual of European descent was paired only once, in which case the order of patients of African descent affected the matchings. Therefore, we generated 10000 random permutations of patients of African descent and assessed the significance of paired comparisons for the matchings based on each permutation.
Statistical analyses were performed with the R software package (www.r-project.org).

Results #1: unadjusted comparisons

- Patients of African descent had slower CD4 decline (n=123 African/463 European; Wilcoxon rank sum test, p<0.001; difference in the median: 24.3 cells/µL/year) and lower viral setpoint (n=143 African/333 European; Wilcoxon rank sum test, p=0.027; difference in the median: 0.22 log10 RNA copies/mL) compared with patients of European descent.
- Subtyping could be performed for 437 patients (80 of African/357 of European descent) with a CD4 slope, and 362 patients (92 of African/270 of European descent) with a setpoint.
- Out of the 4 shared subtypes (A, B, C and CRF 02_AG), patients of African descent had slower CD4 decline in 3, and lower setpoint in all 4.

Testing the Taqman effect:
The Roche Taqman assay has been demonstrated to underestimate virus load, especially with non-B subtypes. We therefore repeated the setpoint analyses on data obtained before the switch to the Taqman assay in the SHCS (2005). This restriction reduced the sample size (96/235), it hardly affected the median setpoint of the two groups, but abolished significance of the difference by Wilcoxon rank sum test (but not by multivariate regression; see Results #2).

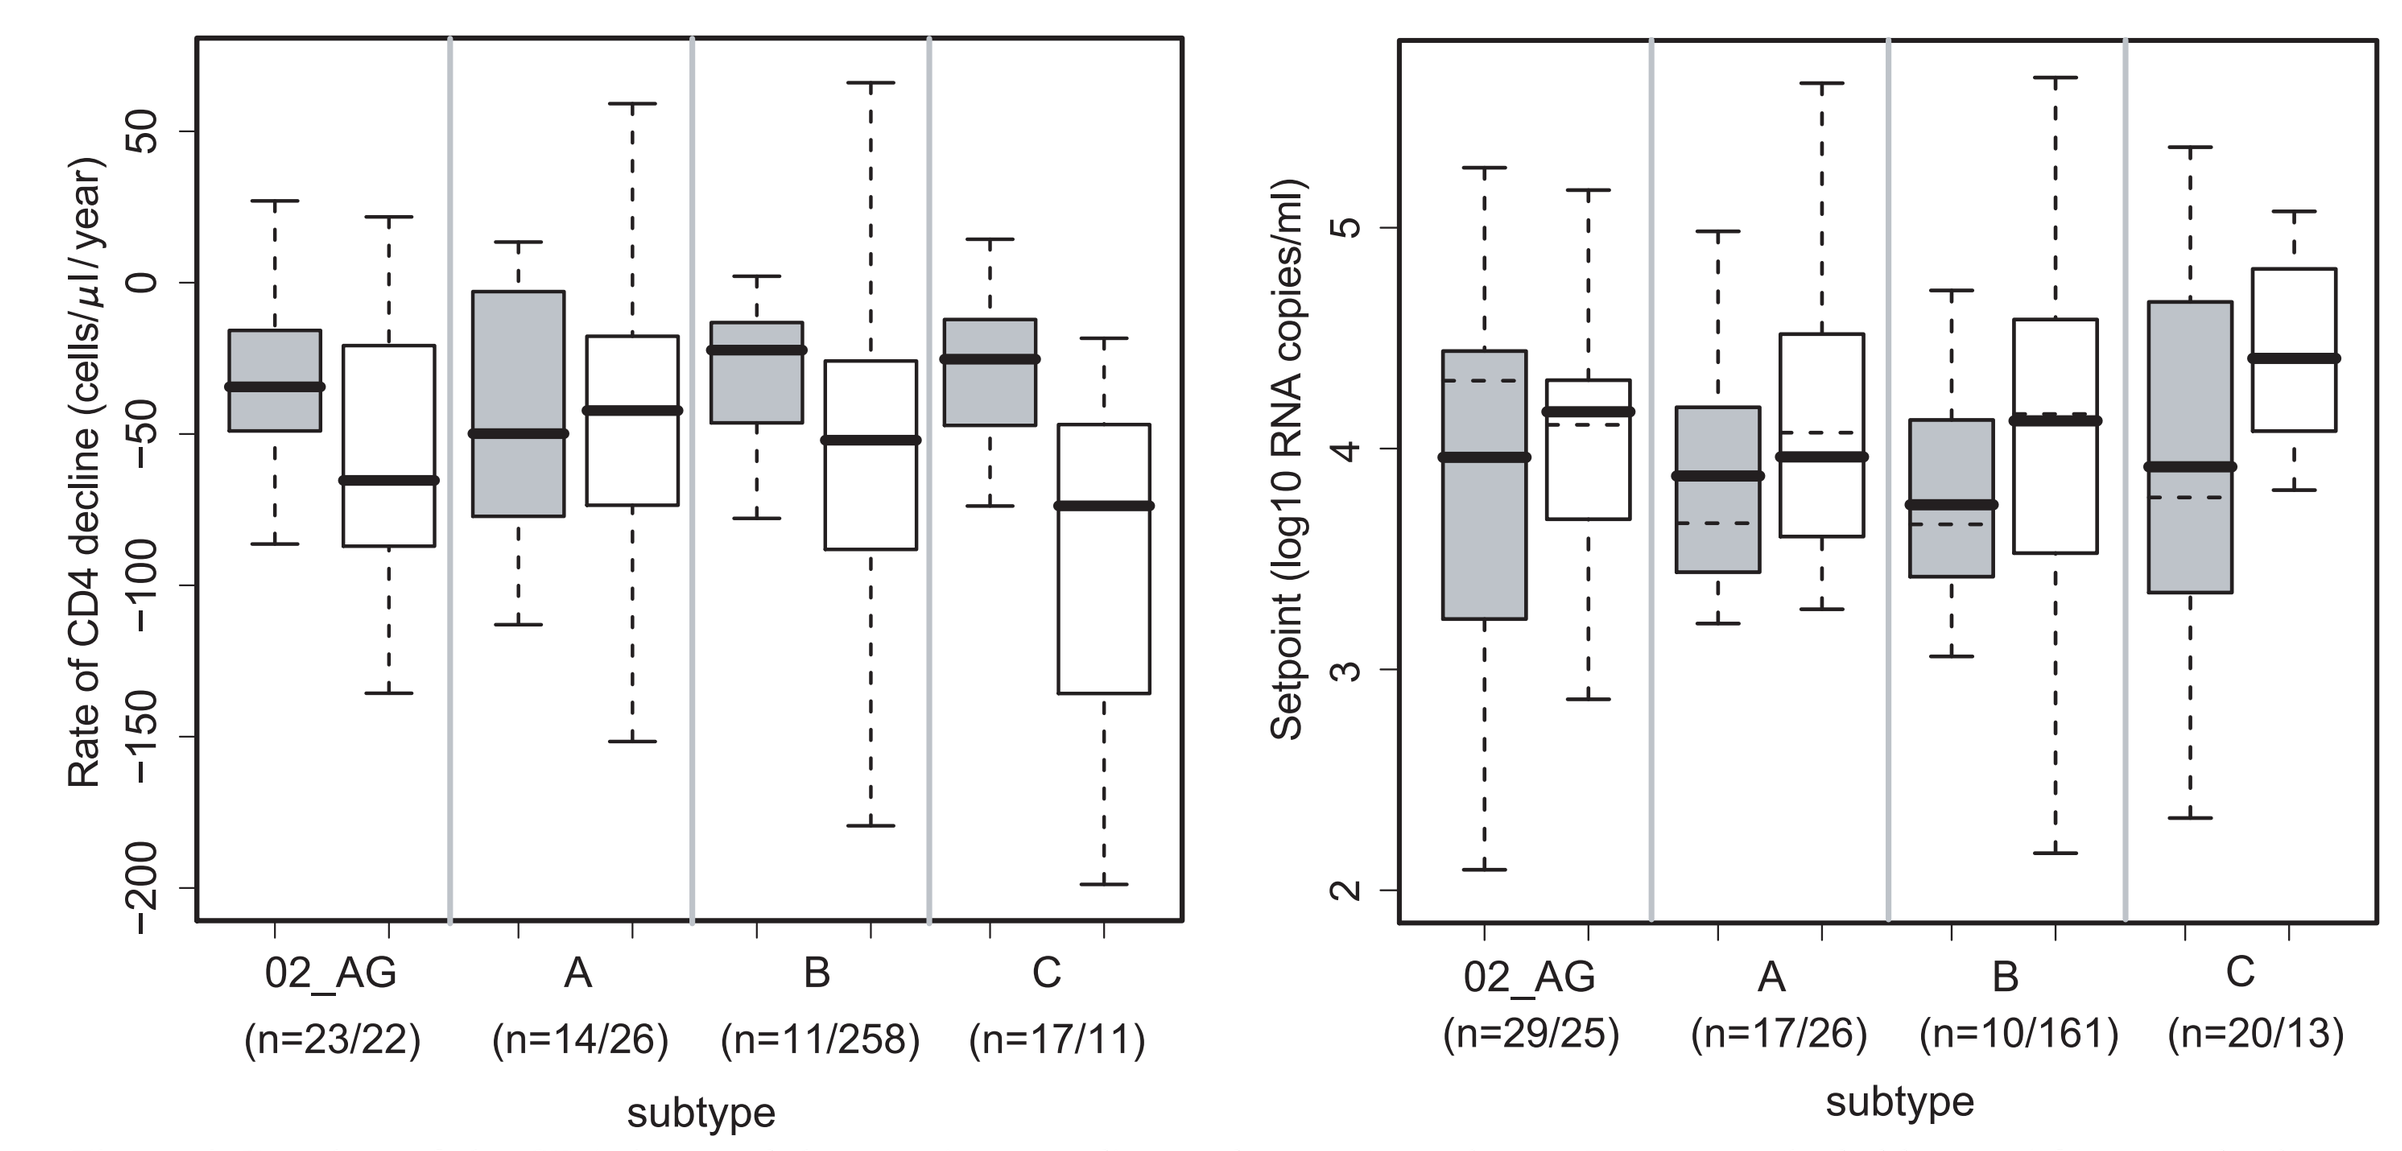
Rate of CD4 decline (cells/µl/year)
Setpoint (log10 RNA copies/ml)
02_AG
(n=23/22)
A
(n=14/26)
B
(n=11/258)
C
(n=17/11)
02_AG
(n=29/25)
A
(n=17/26)
B
(n=10/161)
C
(n=20/13)
subtype
subtype

Figure 1. Boxplots of the CD4 slope and the setpoint according to descent (open boxes: European; shaded boxes: African) and subtype. Boxes indicate interquartile range in each category; median is indicated by horizontal lines within the boxes, and whiskers extend to the farthest values that are not more than 1.5 times the box width away from the box. Sample sizes are shown for both groups (African/European). Median setpoints in the pre-Taqman dataset are shown in dashed lines. CRF 02_AG had higher median in patients of African descent in this restricted data set.

Results #2: controlled comparisons

- Multivariate regression models controlling for gender, subtype, age, date of infection and baseline CD4 count confirmed slower CD4 decline and lower viral setpoint in patients of African descent.
- Effect of subtype(s) was not significant: analyses were repeated including also patients with unknown subtype.

Table 2. The effect of African vs. European descent on markers of disease progression in regression analyses

Regression model (dependent variable)	With subtypes[a]	Without subtypes
Rate of CD4 decline[b]		
n (African/European)	65/317	123/463
Coefficient for African descent[c] (cells/µL/year) [95% CI]	18.0 [2.5–33.5]	26.6 [12.3–41.0]
p	0.02	p<0.001
Viral setpoint[d]		
n (African/European)	76/225	143/333
Coefficient for African descent[c] (log10 RNA copies/mL) [95% CI]	-0.29 [-0.53 – -0.05]	-0.29 [-0.48 – -0.11]
p	0.016	0.001

[a] Included subtypes A, B, C and CRF 02_AG. [b] Additional significant effect was associated with baseline CD4 count.
[c] Difference from patients of European origin (the reference level).
[d] Additional significant effects were associated with gender, baseline CD4 count, date of infection and the interaction between gender and subtype A. The effect of African descent remained significant on pre-Taqman data.

Comparisons matched for baseline CD4 count confirmed slower CD4 decline in patients of African descent:

- Matching with replacement: p<0.001 (sign test); mean difference in the CD4 slopes within pairs: 40.2 cells/µL/year.
- Matching without replacement was performed on 10000 random permutations of the patients of African origin and yielded significant differences (at p<0.01) by sign tests for all permutations, with a median significance p<0.001.

Results #3: CD4 slope vs. viral setpoint

The CD4 slope (primary marker of disease progression) is correlated with the viral setpoint (prognostic marker). We therefore extended the multivariate regression model of the CD4 slope with setpoint as additional control factor.

Table 3. The effect of descent and viral setpoint in multivariate regression of the CD4 slope.

	With subtypes[a]	Without subtypes
n (African/European)	58/168	103/242
Coefficient for African descent (cells/µL/year) [95% CI]	-146 [-223 – -69]	-196 [-284 – -108]
Coefficient for setpoint (European descent) (cells/µL/year/log10 RNA copies/mL) [95% CI]	-42.1 [-53.0 – -31.2]	-46.3 [-55.8 – -36.7]
Coefficient for African descent×setpoint[b] (cells/µL/year/log10 RNA copies/mL) [95% CI]	40.6 [20.9–60.3]	42.3 [25.2–59.3]
Setpoint of descent neutrality[c] (log10 RNA copies/mL)	3.60	4.63

[a] Included subtypes A, B, C and CRF 02_AG. [b] The coefficient for setpoint in Africans is the sum of this interaction term and the coefficient for the European reference level. [c] Setpoint at which the two groups have equal expected CD4 slope when all other factors are kept equal. The results were robust also on pre-Taqman data. All effects were significant at p<0.001.

- The CD4 slope depends strongly on the setpoint in patients of European descent, but not in patients of African descent.

CONCLUSIONS

- **Patients of African descent experience slower CD4 decline and lower viral setpoint compared with patients of European descent in the Swiss HIV Cohort Study.**
- **The effect of descent/ethnicity is independent of the infecting subtypes.**
- **Slower CD4 decline might reflect better tolerance of high virus levels in patients of African descent.**

Hypothesis

Higher levels of antigenic exposure in Africa might have driven adaptation towards lower baseline immune activation. Immune activation is crucial in HIV disease progression. In the low-antigen environment of industrialized countries, individuals of African descent might have lower immune activation compared with individuals of European descent, which might explain the observed differences in HIV disease progression.

Selected references

[1] Müller et al. (2006) Stable virulence levels in the HIV epidemic of Switzerland over two decades. **AIDS**, 20:889-894.
[2] de Oliveira et al. (2005) An automated genotyping system for analysis of HIV-1 and other microbial sequences. **Bioinformatics**, 21:3797-3800.
[3] Myers et al. (2005) A statistical model for HIV-1 sequence classification using the subtype analyser (STAR). **Bioinformatics**, 21:3535-3540.

Acknowledgements

V. M. was supported by the Hungarian Scientific Research Fund (OTKA grant NF72791) and by the European Commission Virolab Project Grant 027446 (www.virolab.org). S. B. acknowledges support by the Swiss National Science Foundation. This study has been financed in the framework of the Swiss HIV Cohort Study, supported by the Swiss National Science Foundation (3345–062041). Further support was provided by Swiss National Science Foundation grant 3247B0–112594 (to H.F.G., S.Y., S.B. and B.L.), SHCS projects 470, 528, the SHCS research foundation, and by a further research grant of the Union Bank of Switzerland in the name of a donor (to H.F.G.).
The members of the Swiss HIV Cohort Study are M. Battegay, E. Bernasconi, J. Böni, HC Bucher, Ph. Bürgisser, A. Calmy, S. Cattacin, M. Cavassini, R. Dubs, M. Egger, L. Elzi, M. Fischer, M. Flepp, A. Fontana, P. Francioli (President of the SHCS, Centre Hospitalier Universitaire Vaudois, CH-1011- Lausanne), H. Furrer (Chairman of the Clinical and Laboratory Committee), C. Fux, M. Gorgievski, H. Günthard (Chairman of the Scientific Board), H. Hirsch, B. Hirschel, I. Hösli, Ch. Kahlert, L. Kaiser, U. Karrer, C. Kind, Th. Klimkait, B. Ledergerber, G. Martinetti, B. Martinez, N. Müller, D. Nadal, M. Opravil, F. Paccaud, G. Pantaleo, A. Rauch, S. Regenass, M. Rickenbach (Head of Data Center), C. Rudin (Chairman of the Mother & Child Substudy), P. Schmid, D. Schultze, J. Schüpbach, R. Speck, P. Taffé, A. Telenti, A. Trkola, P. Vernazza, R. Weber, S. Yerly.
We thank all patients participating in the Swiss HIV Cohort Study, all doctors, study nurses and laboratory technicians involved, and the datacenter. Furthermore, we are grateful to technicians of the SHCS resistance laboratories for the high quality of sequence data and thank Smartgene for permanent service and support.